5935 - A0432

MESOPIC VISUAL FUNCTION ATTAINED BY **LENSES SPECIFICALLY DESIGNED FOR NIGHT DRIVING**

Chamorro, E, Cleva, JM*, Álvarez, M*, Subero, M*, Garcia-Ortega, M*, Alonso, J**
**Indizen Optical Technologies S.L., Spain*

Corresponding author Eva Chamorro

ARVO

INTRODUCTION

Driving is a complex task that requires specific visual skills and optimal vision to clearly see and react to traffic signals, traffic lights, pedestrians and other vehicles. The **nighttime driving environment presents challenging visual conditions for drivers**, including dim street lighting, glare from oncoming headlights, and the need to quickly adapt across a wide range of lighting levels, inducing changes in visual perception such as loss of visual acuity, increased glare or night myopia.

Also, fixation changes during day and night driving conditions. At nighttime, the area of fixation is smaller than in daytime driving and the eye tends to fixate lower.
According to the study lead by Brimley et al., these differences can be attributed to reduced visibility and reliance on headlamps at night *(Figure 1)*.

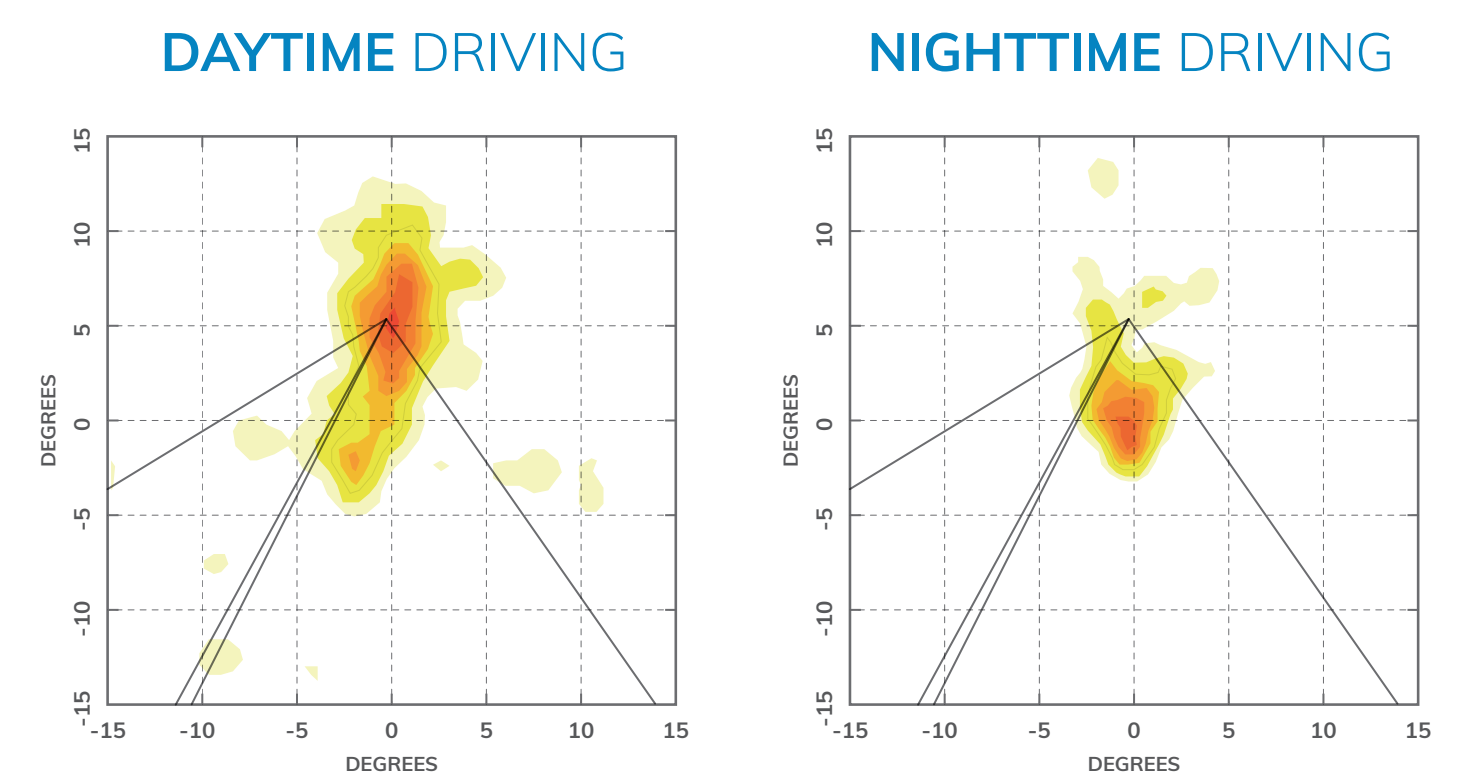
DAYTIME DRIVING
NIGHTTIME DRIVING
DEGREES
DEGREES
-15
-10
-5
0
5
10
15

Figure 1: Gaze fixation maps of different driving conditions according to the Brimley study[3]

Recently, new single vision and progressive power lenses (PPL)(inMotion™, IOT) have been proposed to improve day and night driving. These lenses have **variations in the progression profile of the lens, providing a slight negative defocus in a small region just above the pupil. The power distribution maps are also optimized for distance vision and a special blue light filter was included in the lens to enhance night driving vision** *(Figure 2)*.

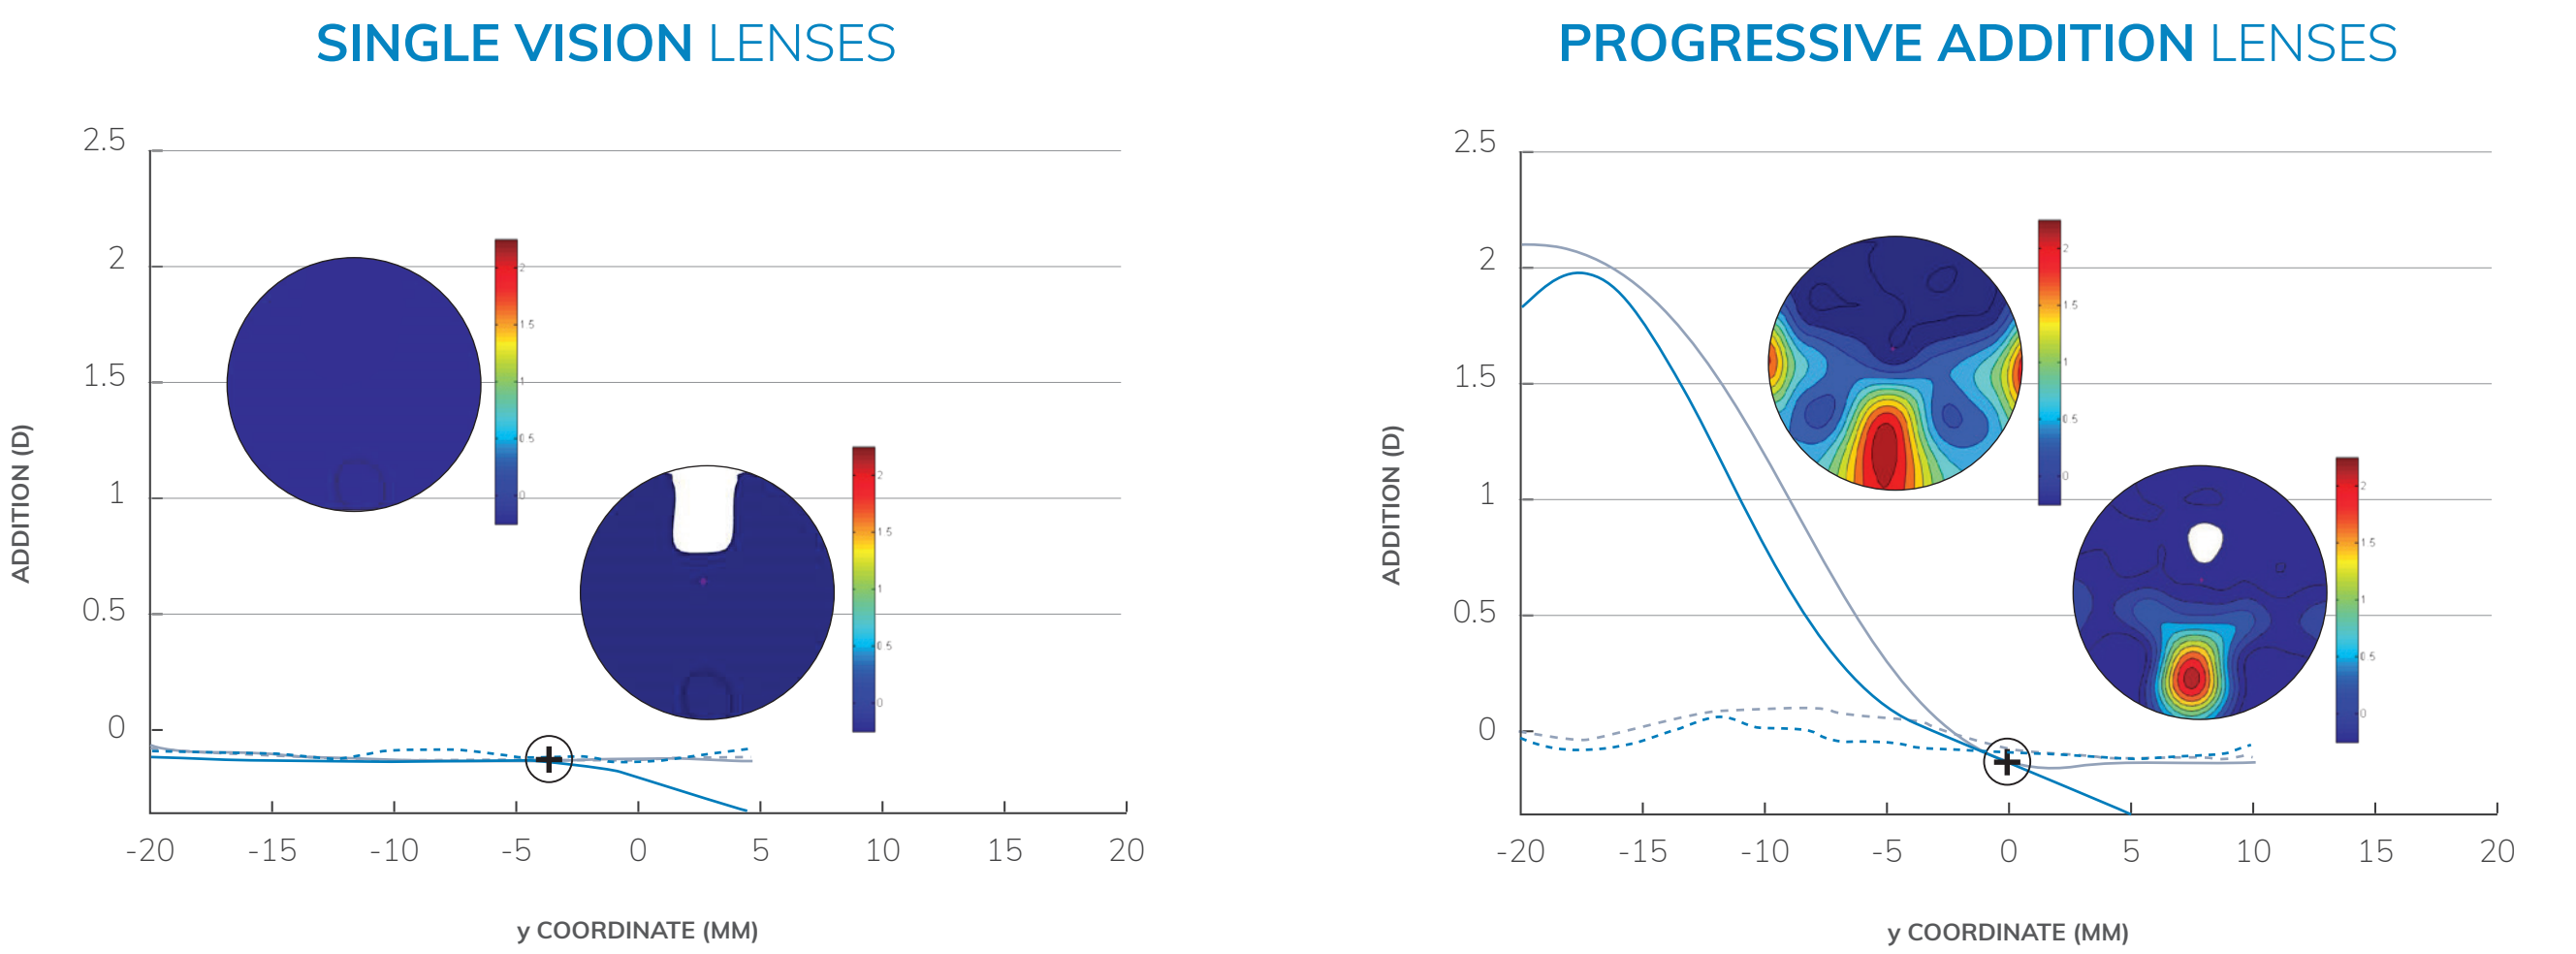
SINGLE VISION LENSES
PROGRESSIVE ADDITION LENSES
ADDITION (D)
2.5
2
1.5
1
0.5
0
-20
-15
-10
-5
0
5
10
15
20
y COORDINATE (MM)

Figure 2: Progression profile and mean power color maps for the lenses proposed to improve mesopic vision in comparison to standard lenses.

MESOPIC VISUAL FUNCTION ATTAINED BY **LENSES SPECIFICALLY DESIGNED FOR NIGHT DRIVING**

5935 - A0432

IOT

ARVO

Chamorro, E, Cleva, JM*, Álvarez, M*, Subero, M*, Garcia-Ortega, M*, Alonso, J**
**Indizen Optical Technologies S.L., Spain*

Corresponding author Eva Chamorro

PURPOSE

The goal of this study is to compare the mesopic visual function of users wearing general-use lenses and lenses optimized for mesopic vision.

METHOD

56 subjects participated in this observational, prospective and double-blind trial. **Visual function in mesopic illumination conditions after 15 minutes of dark adaptation was evaluated** while users were wearing **2 different lenses**:

- **Control lens**: General-use design with standard power distribution and standard anti-reflective coating.

- **Test lens**: Special design developed ad-hoc for this study and characterized by a region with a slight negative defocus above the fitting cross and a special blue light filter.

Participants under 45 years of age tested single vision lenses (group 1, n=26) and subjects over 45 years of age tested progressive lenses (group 2, n=30). The parameters analyzed were low-contrast undistorted distance visual field using a rotational platform *(Figure 3)*, glare recovery time after flash, subjective satisfaction (scale 1-5) after participants used both pairs of lenses for 7 days, and preferred design for driving. Statistical analysis was performed using *Statgraphics Centurion XVI.II* software.

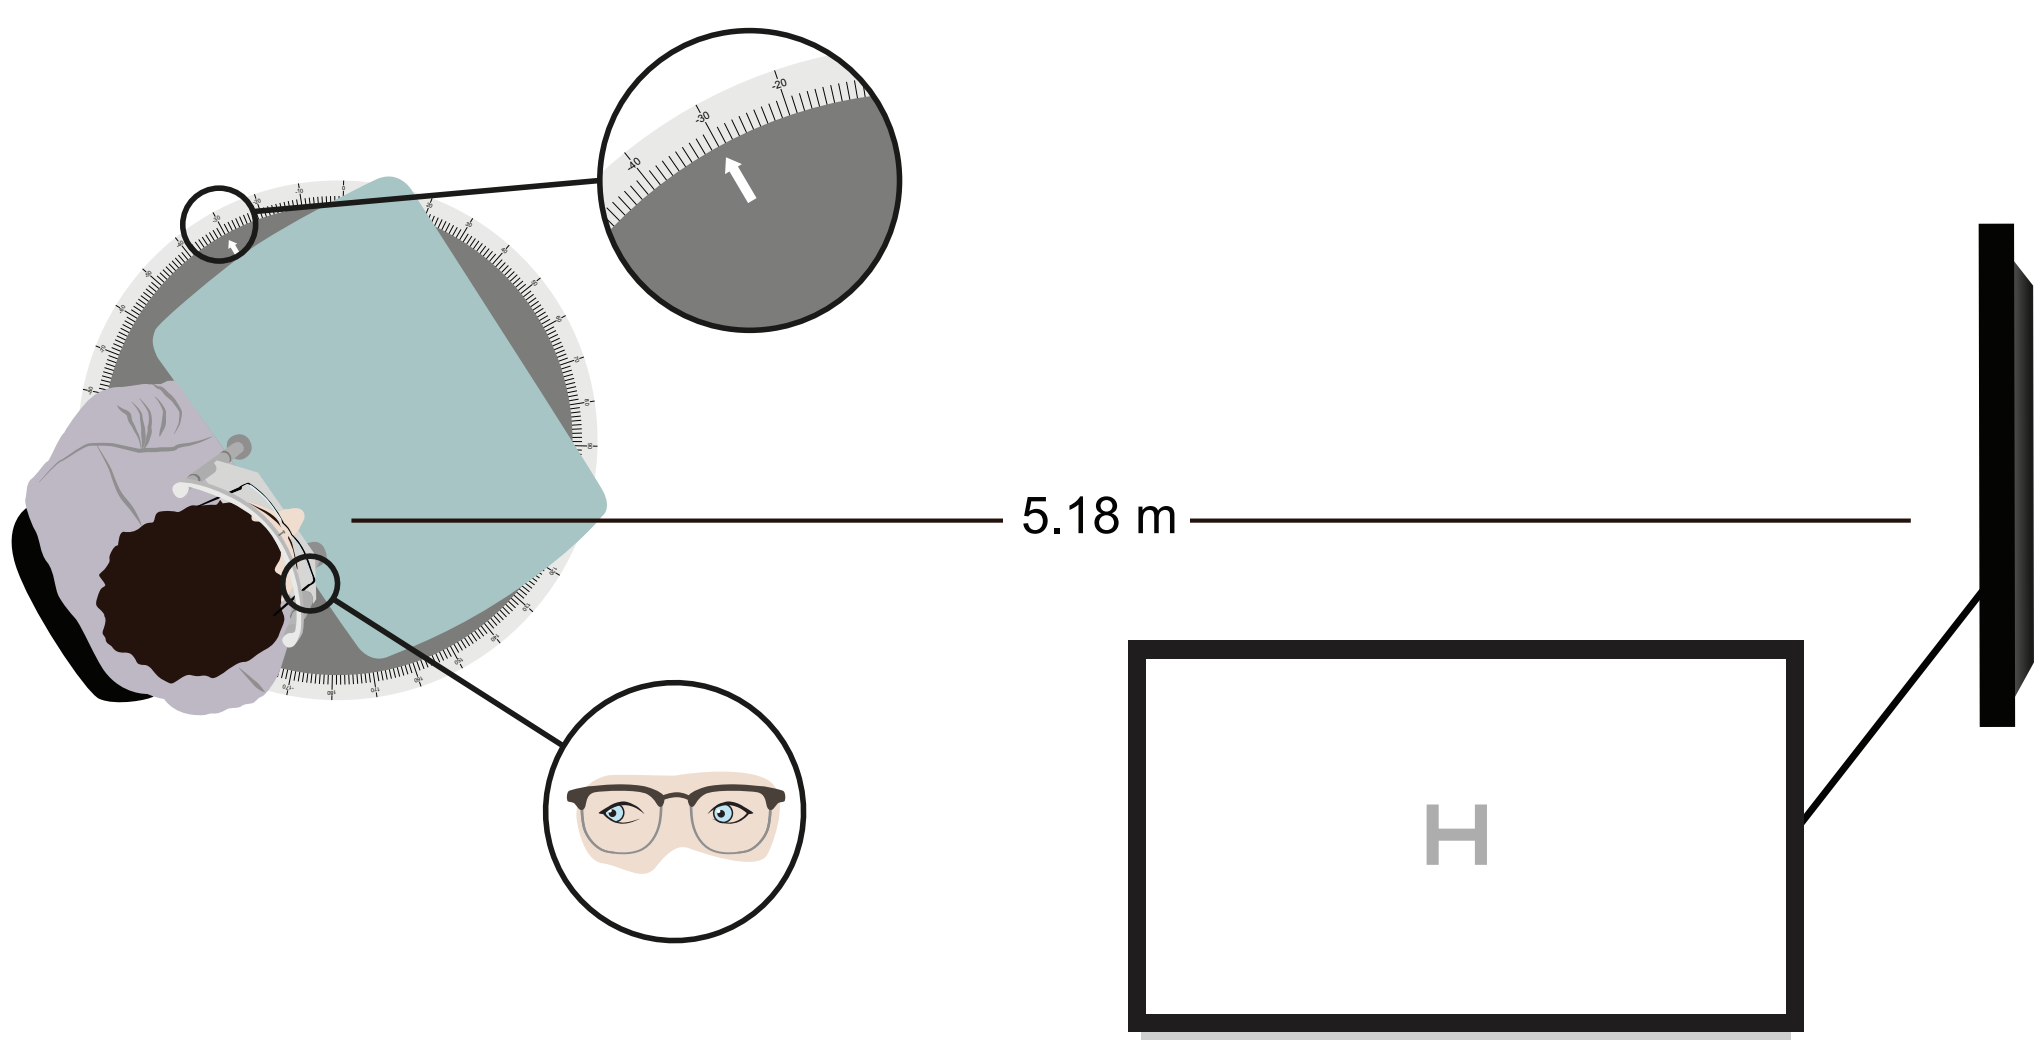
5.18 m
H

Figure 3: Method for evaluation of low-contrast undistorted visual field.

MESOPIC VISUAL FUNCTION ATTAINED BY **LENSES SPECIFICALLY DESIGNED FOR NIGHT DRIVING**

5935 - A0432

ARVO

Chamorro, E, Cleva, JM*, Álvarez, M*, Subero, M*, Garcia-Ortega, M*, Alonso, J**
**Indizen Optical Technologies S.L., Spain*

Corresponding author Eva Chamorro

RESULTS

A lower glare recovery time after flash as well as a significant higher satisfaction was observed for group 2 (people over 45) with the special design. For both groups, low-contrast and an undistorted visual field was significantly wider with the special design. The specially designed lens for mesopic vision was preferred for driving in both age groups (*Figure 4*).

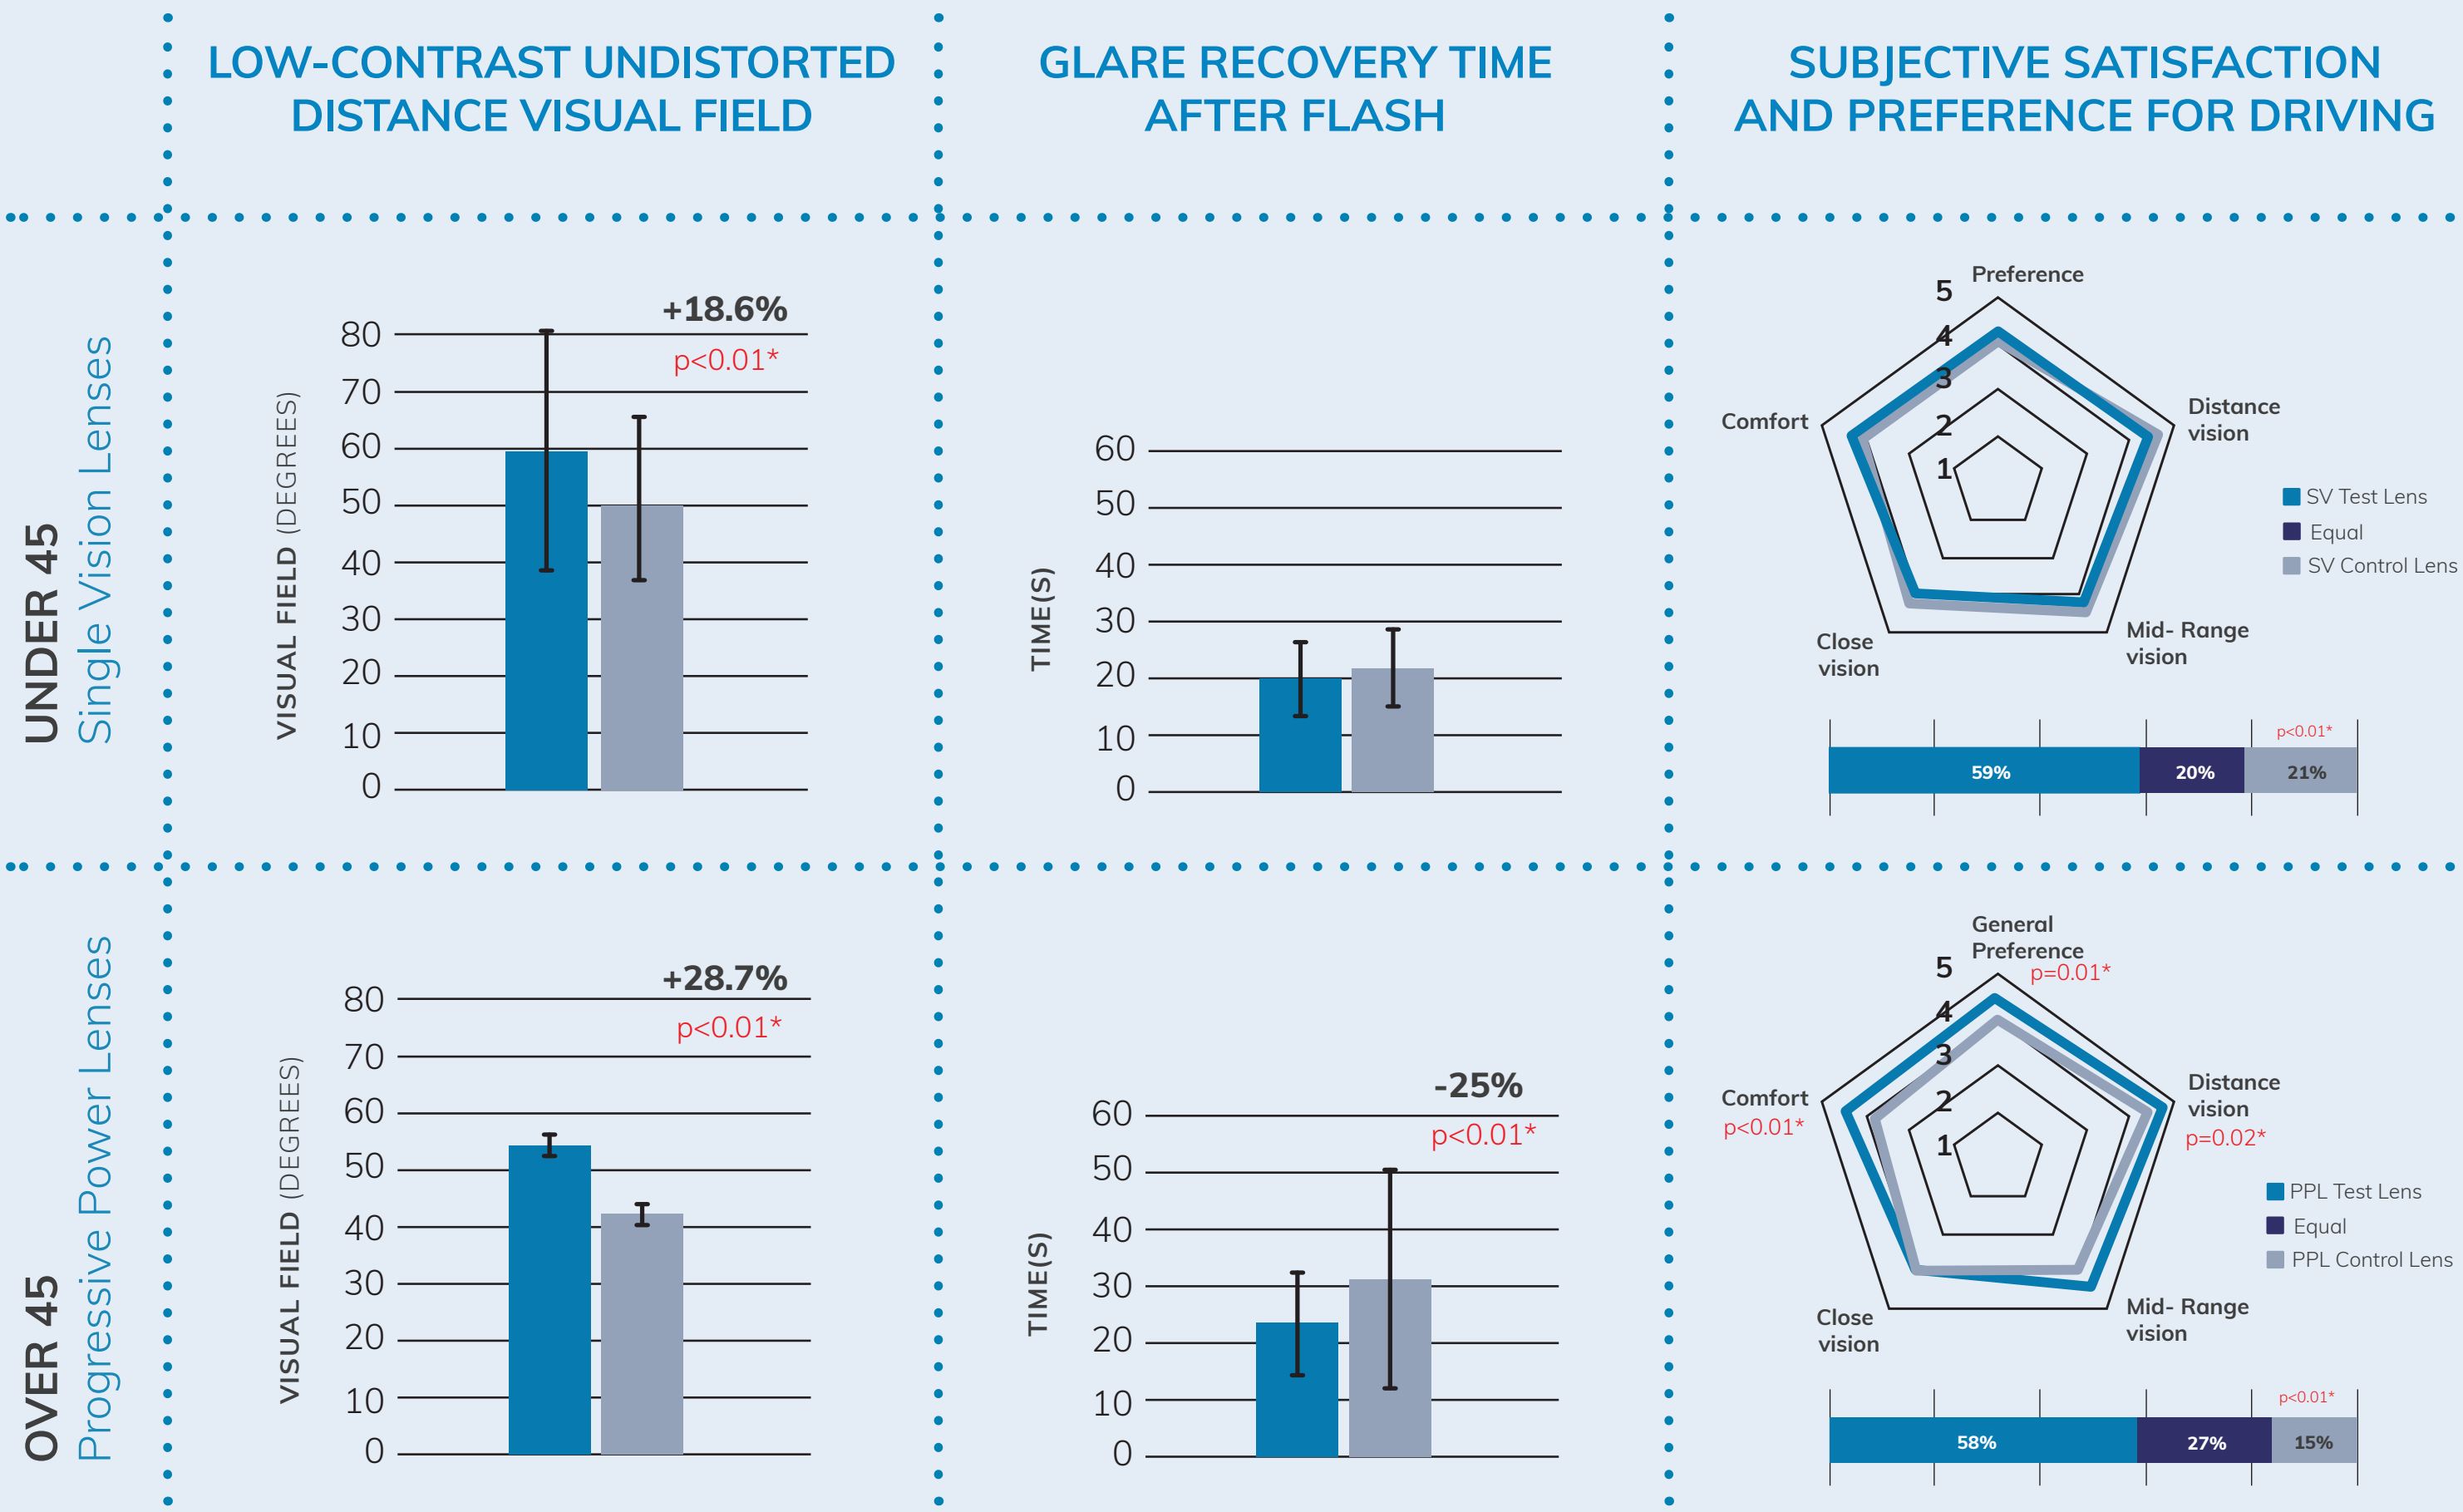
LOW-CONTRAST UNDISTORTED DISTANCE VISUAL FIELD
GLARE RECOVERY TIME AFTER FLASH
SUBJECTIVE SATISFACTION AND PREFERENCE FOR DRIVING
UNDER 45
Single Vision Lenses
VISUAL FIELD (DEGREES)
+18.6%
p<0.01*
TIME(S)
Preference
Distance vision
Mid- Range vision
Close vision
Comfort
SV Test Lens
Equal
SV Control Lens
p<0.01*
59%
20%
21%
OVER 45
Progressive Power Lenses
VISUAL FIELD (DEGREES)
+28.7%
p<0.01*
TIME(S)
-25%
p<0.01*
General Preference
p=0.01*
Distance vision
p=0.02*
Comfort
p<0.01*
Mid- Range vision
Close vision
PPL Test Lens
Equal
PPL Control Lens
p<0.01*
58%
27%
15%

Figure 4: Clinical assessment of control and test lenses.

CONCLUSIONS

A new lens-design concept characterized by special power distribution maps and incorporating a blue light filter, improved mesopic visual function. These lenses provided a wider undistorted distance visual field, lower glare recovery time, and higher user satisfaction in general, and specifically for driving.

5935 - A0432

MESOPIC VISUAL FUNCTION ATTAINED BY **LENSES SPECIFICALLY DESIGNED FOR NIGHT DRIVING**

Chamorro, E, Cleva, JM*, Álvarez, M*, Subero, M*, Garcia-Ortega, M*, Alonso, J**
**Indizen Optical Technologies S.L., Spain*

Corresponding author Eva Chamorro

REFERENCES

1. Mestre DR. Visual factors in driving. In: Fuller R, Santos JA (eds), Human factors for highway engineers. Amsterdam; New York: Pergamon; 2002.
2. Serafin C. Driver Eye Fixations On Rural Roads: Insight Into Safe Driving Behavior. Interim Report. University of Michigan Transportation Research Institute; 1994.
3. Brimley BK, Carlson PJ, Hawkins HG. Use of Fixation Heat Maps to Evaluate Visual Behavior of Unfamiliar Drivers on Horizontal Curves. Transport Res Rec 2014;16-26.
4. Chamorro E, Cleva J, Concepcion P, Subero MS, Alonso J. Lens Design Techniques to Improve Satisfaction in Free-Form Progressive Addition Lens Users. JOJ Ophthalmology 2018;6:1-10.
5. Concepcion P, Cleva J, Chamorro E, et al. Clinical Evaluation of Free-Form PALs: Customized vs Standard Designs. European Academy of Optometry and Optics annual conference. Barcelona, Spain; 2017.

inMotion is a trademark of Indizen Optical Technologies S.L.